Discovery of novel 5-HT2A receptor agonists with psychedelic drug-like in vitro and in vivo pharmacological activity and therapeutic potential for treatment-resistant depression

Carrie A. Bowen, Tanweer A. Khan, Robert B. Perni, Rebecca Aron, Alan Gibbs, Rick Shin, Adam L. Halberstadt, Isabella Premoli, Patrick Kleine, **Srinivas Rao***, and Glenn F. Short III

atai LIFE SCIENCES

EntheogeniX

2023 ACNP Annual Meeting
Poster Session II
Poster #: T150

Background/Objective

Mood disorders, particularly treatment-resistant depression (TRD), remain a significant unmet medical need [1]. Clinical and preclinical research suggest the potential for serotonergic psychedelic compounds, such as the tryptamine psilocybin, to produce rapid and long-lasting antidepressant activity after a single dose [2-3].

The goal of the current research was to discover novel 5-HT2A receptor (5-HT2AR) agonists with optimal drug-like properties showing in vitro and in vivo pharmacological profiles similar to hallucinogenic tryptamines. As many known psychedelic molecules exhibit poor selectivity across serotonin receptors, we focused on novel agonists with improved selectivity for 5-HT2AR over 5-HT2B receptor (5-HT2BR) to potentially limit the risk of physiological effects such as valvular heart disease, which can occur in humans after prolonged 5-HT2BR stimulation.

Artificial intelligence/machine learning-driven de novo drug design, followed by medicinal chemistry structure-activity relationship development, was used to discover novel small molecules with selective agonist activity at 5-HT2AR over 5-HT2BR.

In vitro pharmacological characterization of novel compounds was performed using cell-based assays of human 5-HT2AR and 5-HT2BR agonism.

In vivo pharmacological testing included the mouse head twitch response (HTR) assay as a behavioral proxy for the psychedelic potential of 5-HT2AR agonists, due to the robust correlation between the HTR in mice and psychedelic effects in humans for a large series of 5-HT2AR agonists [4]. Antidepressant drug-like activity was assessed using the rat forced swim test (FST) and translational electroencephalography (EEG)-based measures of rapid eye movement (REM) sleep, an increase of which is a hallmark of depression in humans that is suppressed by acute and chronic antidepressant treatment [5-7]. As Wistar-Kyoto (WKY) rats exhibit increased REM sleep that is resistant to suppression by traditional antidepressant drugs (e.g., SSRIs) [8], these animals were employed as a translational model of TRD.

Methods

Human 5-HT2AR Calcium Flux Assay: Gq-mediated secondary messenger signaling of calcium mobilization was monitored with a calcium-sensitive dye and was used as a readout for GPCR activation. Stably-transfected cell lines (U2OS) expressing human 5-HT2AR were loaded with the calcium-sensitive dye in exchange of culture media prior to treatments. Reference and test compound agonist activity was measured on a FLIPR Tetra (MDS) via fluorescence detection of the calcium-sensitive dye. 5-HT was used as the assay reference agonist. Data were normalized to the maximal and minimal response observed in the presence of control ligand and vehicle.

Human 5-HT2AR and 5-HT2BR IPOne Assays: Using stably-transfected cell lines (CHO-K1) expressing human 5-HT2AR or 5-HT2BR, activation of Gq-mediated secondary messenger signaling of myo-Inositol 1 phosphate (IP1) production was detected by a Homogeneous Time-Resolved Fluorescence (HTRF) competitive immunoassay, whereby an IP1 analog coupled to a fluorophore (acceptor) competes with endogenous IP1 for binding to a labeled anti-IP1 antibody (donor). The resulting signal was inversely proportional to the concentration of IP1 in the sample. Cells were incubated with an IP1 inhibitor (to prevent degradation and allow detection) and either reference or test compound. α-Me-5-HT was used as the assay reference agonist. Activation of 5-HT2AR or 5-HT2BR was measured via accumulation of IP1 detected by HTRF. Agonist activity of test compounds was expressed as a percentage of the activity of the reference agonist at its EC100 concentration.

Mouse HTR: Male C57BL/6J mice were implanted with a small neodymium magnet fixed to the cranium with dental cement. On test days, adult mice (n=5-6/group) were injected intraperitoneally (IP) with vehicle negative control or EGX-A (0.3, 1, 3, 10, 30 mg/kg IP prepared in 40% (w/v) hydroxypropyl-b-cyclodextrin in water), immediately placed in a glass cylinder surrounded by a magnetometer coil, and activity was recorded for 30 min. In an antagonism study, the 5-HT2AR antagonist, M100907 (0.001, 0.01, 0.1 mg/kg IP) was injected 20 min prior to EGX-A. Coil voltage was amplified, low pass filtered (2 kHz cutoff), and digitized (20 kHz sampling rate). HTR were identified in the recordings using a validated technique based on artificial intelligence [9]. Mean number of HTR recorded during the test was analyzed using a 1-way Analysis of Variance (ANOVA) followed by Dunnett's test to compare test conditions to the vehicle group. The level of significance was set at $p<0.05$.

Rat FST: This assay measures immobility behavior that follows an inability to escape from a cylinder of water. Traditional and novel antidepressant drugs reduce immobility in the test. Adult male Sprague-Dawley (SD) rats were handled daily for up to 1 week prior to dosing. On Day 1, following a 1-hour acclimation to the testing room, rats were subjected to a 15 min pre-swim in 24 ± 1°C water. Once dried, rats (n=10/group) were administered ketamine positive control (10 mg/kg IP, dosed at 23.5h before FST) or vehicle or EGX-A (3, 10, 30 mg/kg IP, dosed at 23.5, 16, and 0.5h before FST). Data reflect the mean frequency of immobility behavior recorded during the test. A 1-way ANOVA followed by Dunnett's test was used to compare test conditions to the vehicle group. The level of significance was set at $p<0.05$.

WKY Rat EEG: Male WKY rats (n=7) were implanted with a surface EEG dipole electrode (frontal positive, occipital negative), EMG electrodes (nuchal muscle) and an IP radio transmitter to wirelessly transmit signals. On test days, 1 hour after lights-on, adult rats were placed individually in recording boxes similar to their home cages (with food, water and limited environmental enrichment). After 1 hour of habituation, each rat was briefly removed from its box to be dosed and then recordings continued uninterrupted for 23 hours. All rats received all treatments in a pseudo randomized cross-over design with a minimum of 7 days washout between doses. Each treatment condition was represented during each weekly test session, including psilocybin positive control (10 mg/kg IP), vehicle or EGX-A (3, 10, 30 mg/kg IP). Automatic scoring of REM sleep was performed on 10-second epochs of EEG/EMG recordings using proprietary software. REM sleep latency was defined as the time after dosing at which the first 3 consecutive REM sleep epochs occurred. Compound effects on the percentage of time spent in REM sleep were analyzed over the initial 6 hours of EEG recording post-dosing. Data were analyzed by 1- or 2-way repeated measures ANOVA followed by Dunnett's test to compare test conditions to the vehicle group. The level of significance was set at $p<0.05$.

Compound In Vitro 5-HT2AR/ 5-HT2BR Profiles

Figure 1. Analysis of structure-activity relationships identified highly active 5-HT2AR agonists, starting from an initial hit (EGX-1) to EGX compounds with activity greater than psilocin, the active metabolite of psilocybin.

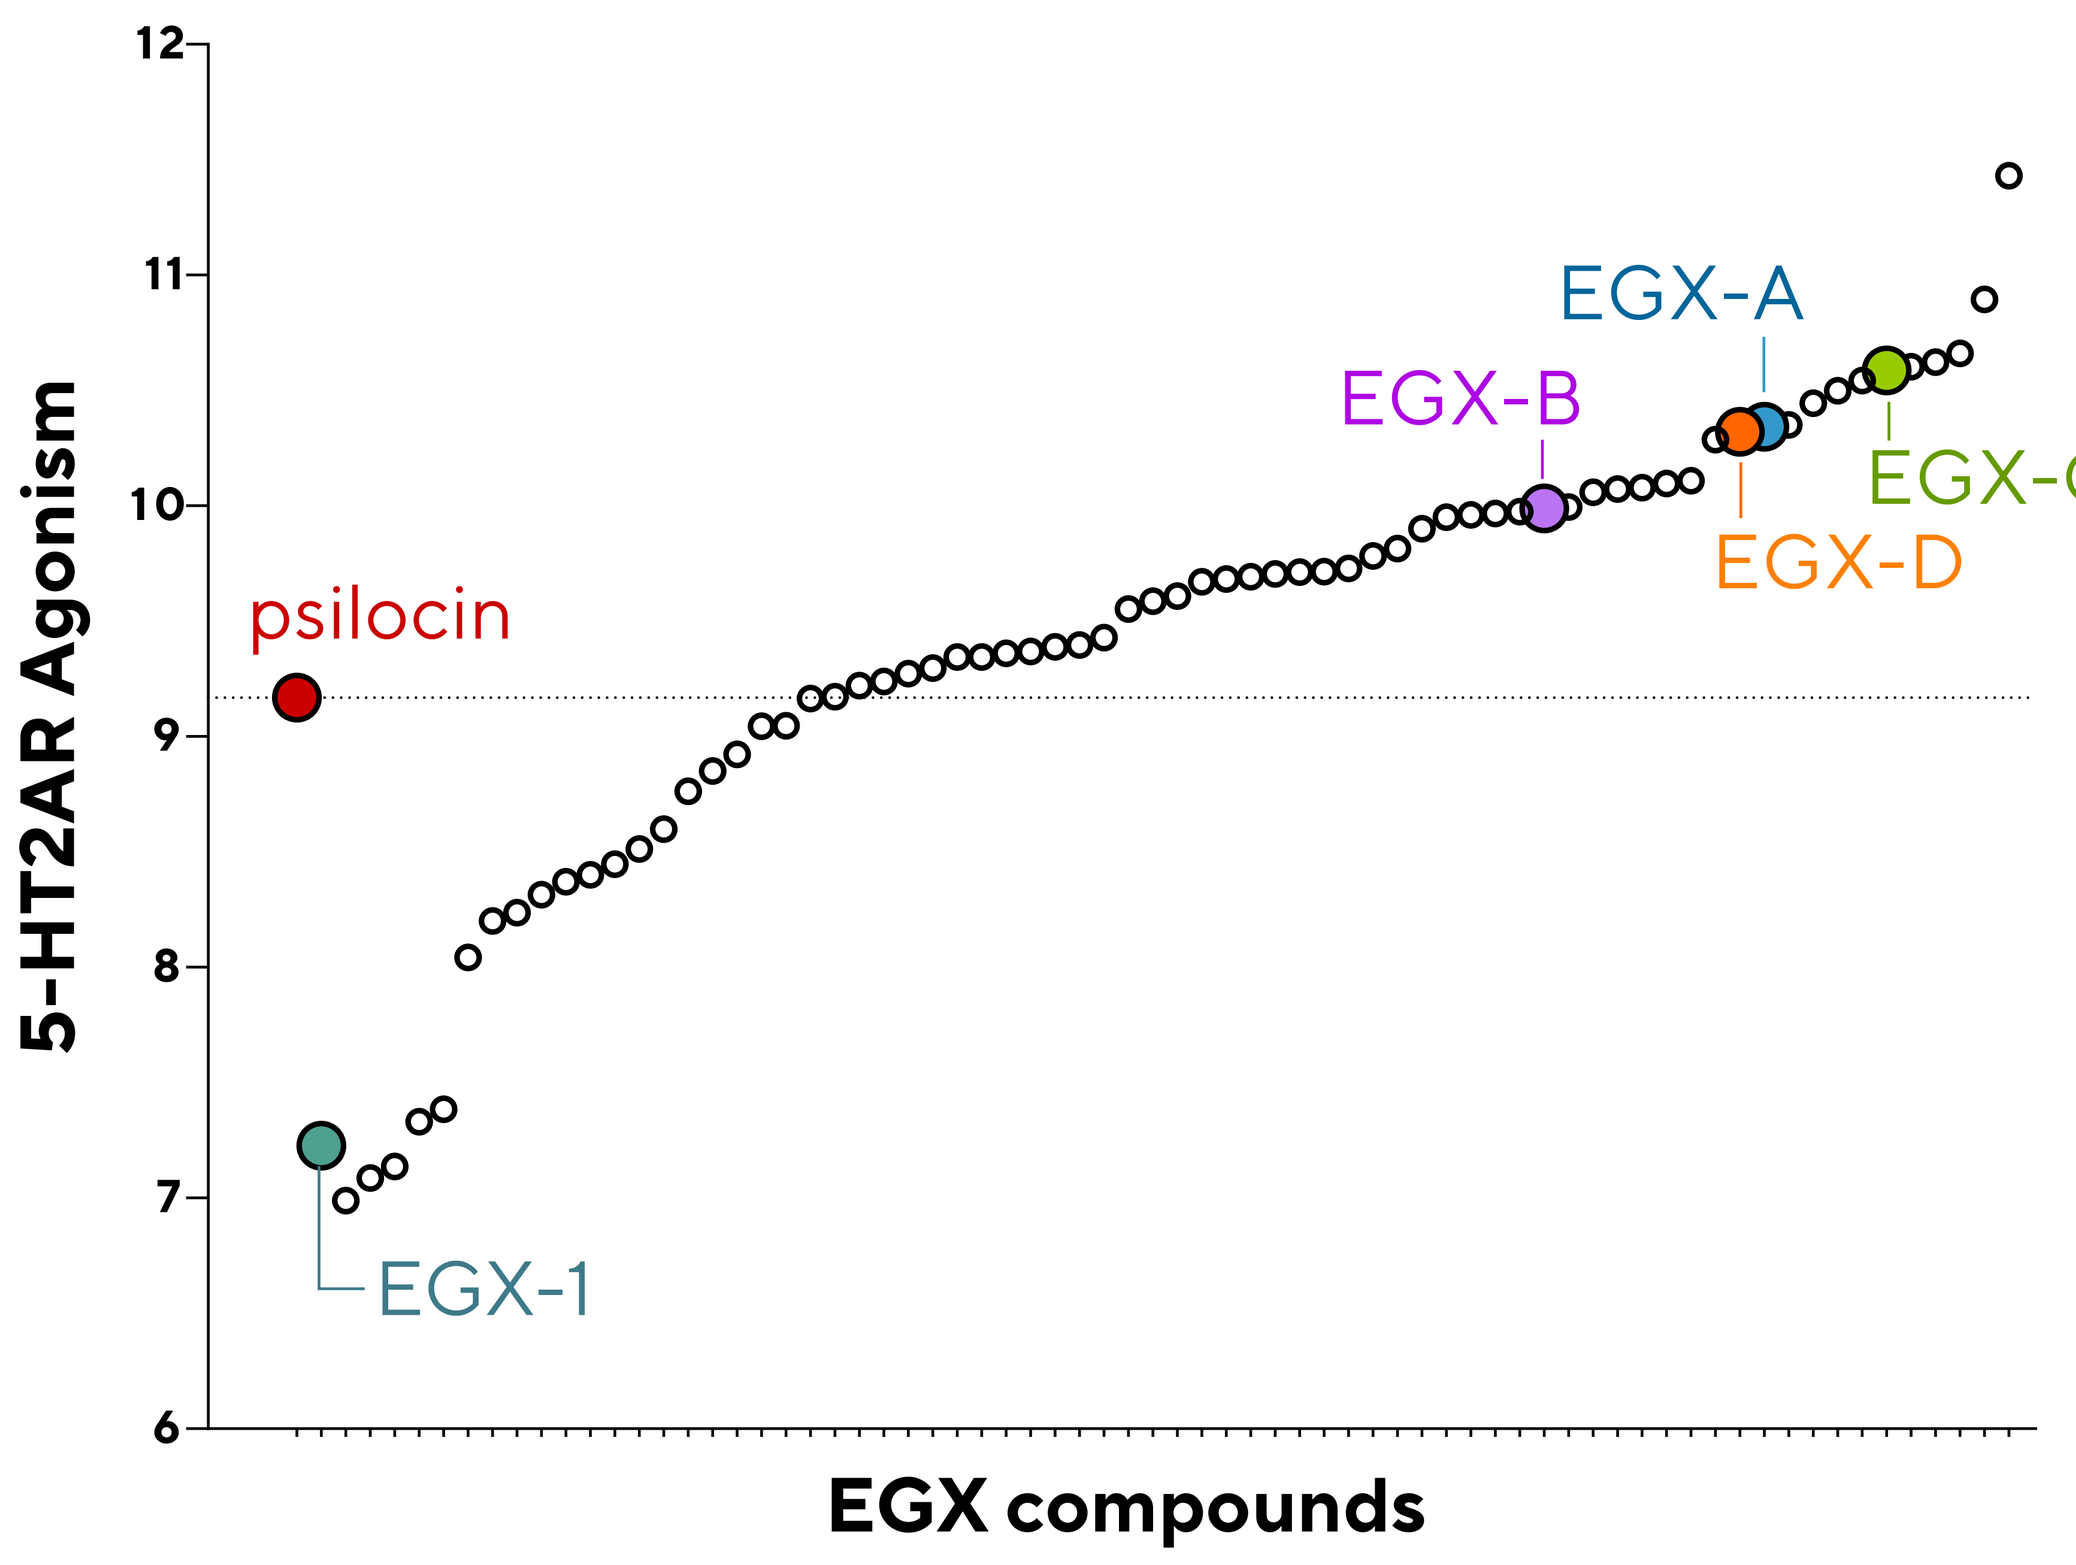
5-HT2AR Agonism
12
11
10
9
8
7
6
psilocin
EGX-1
EGX-A
EGX-B
EGX-C
EGX-D
EGX compounds

Figure 1. Novel EGX compounds rank-ordered by 5-HT2AR agonist activity (log(Emax/EC50), calcium flux assay) with increasing agonist activity from left to right.

Figure 2. Discovery of novel compounds that show agonist selectivity for 5-HT2AR over 5-HT2BR, including EGX compounds with greater selectivity than psilocin.

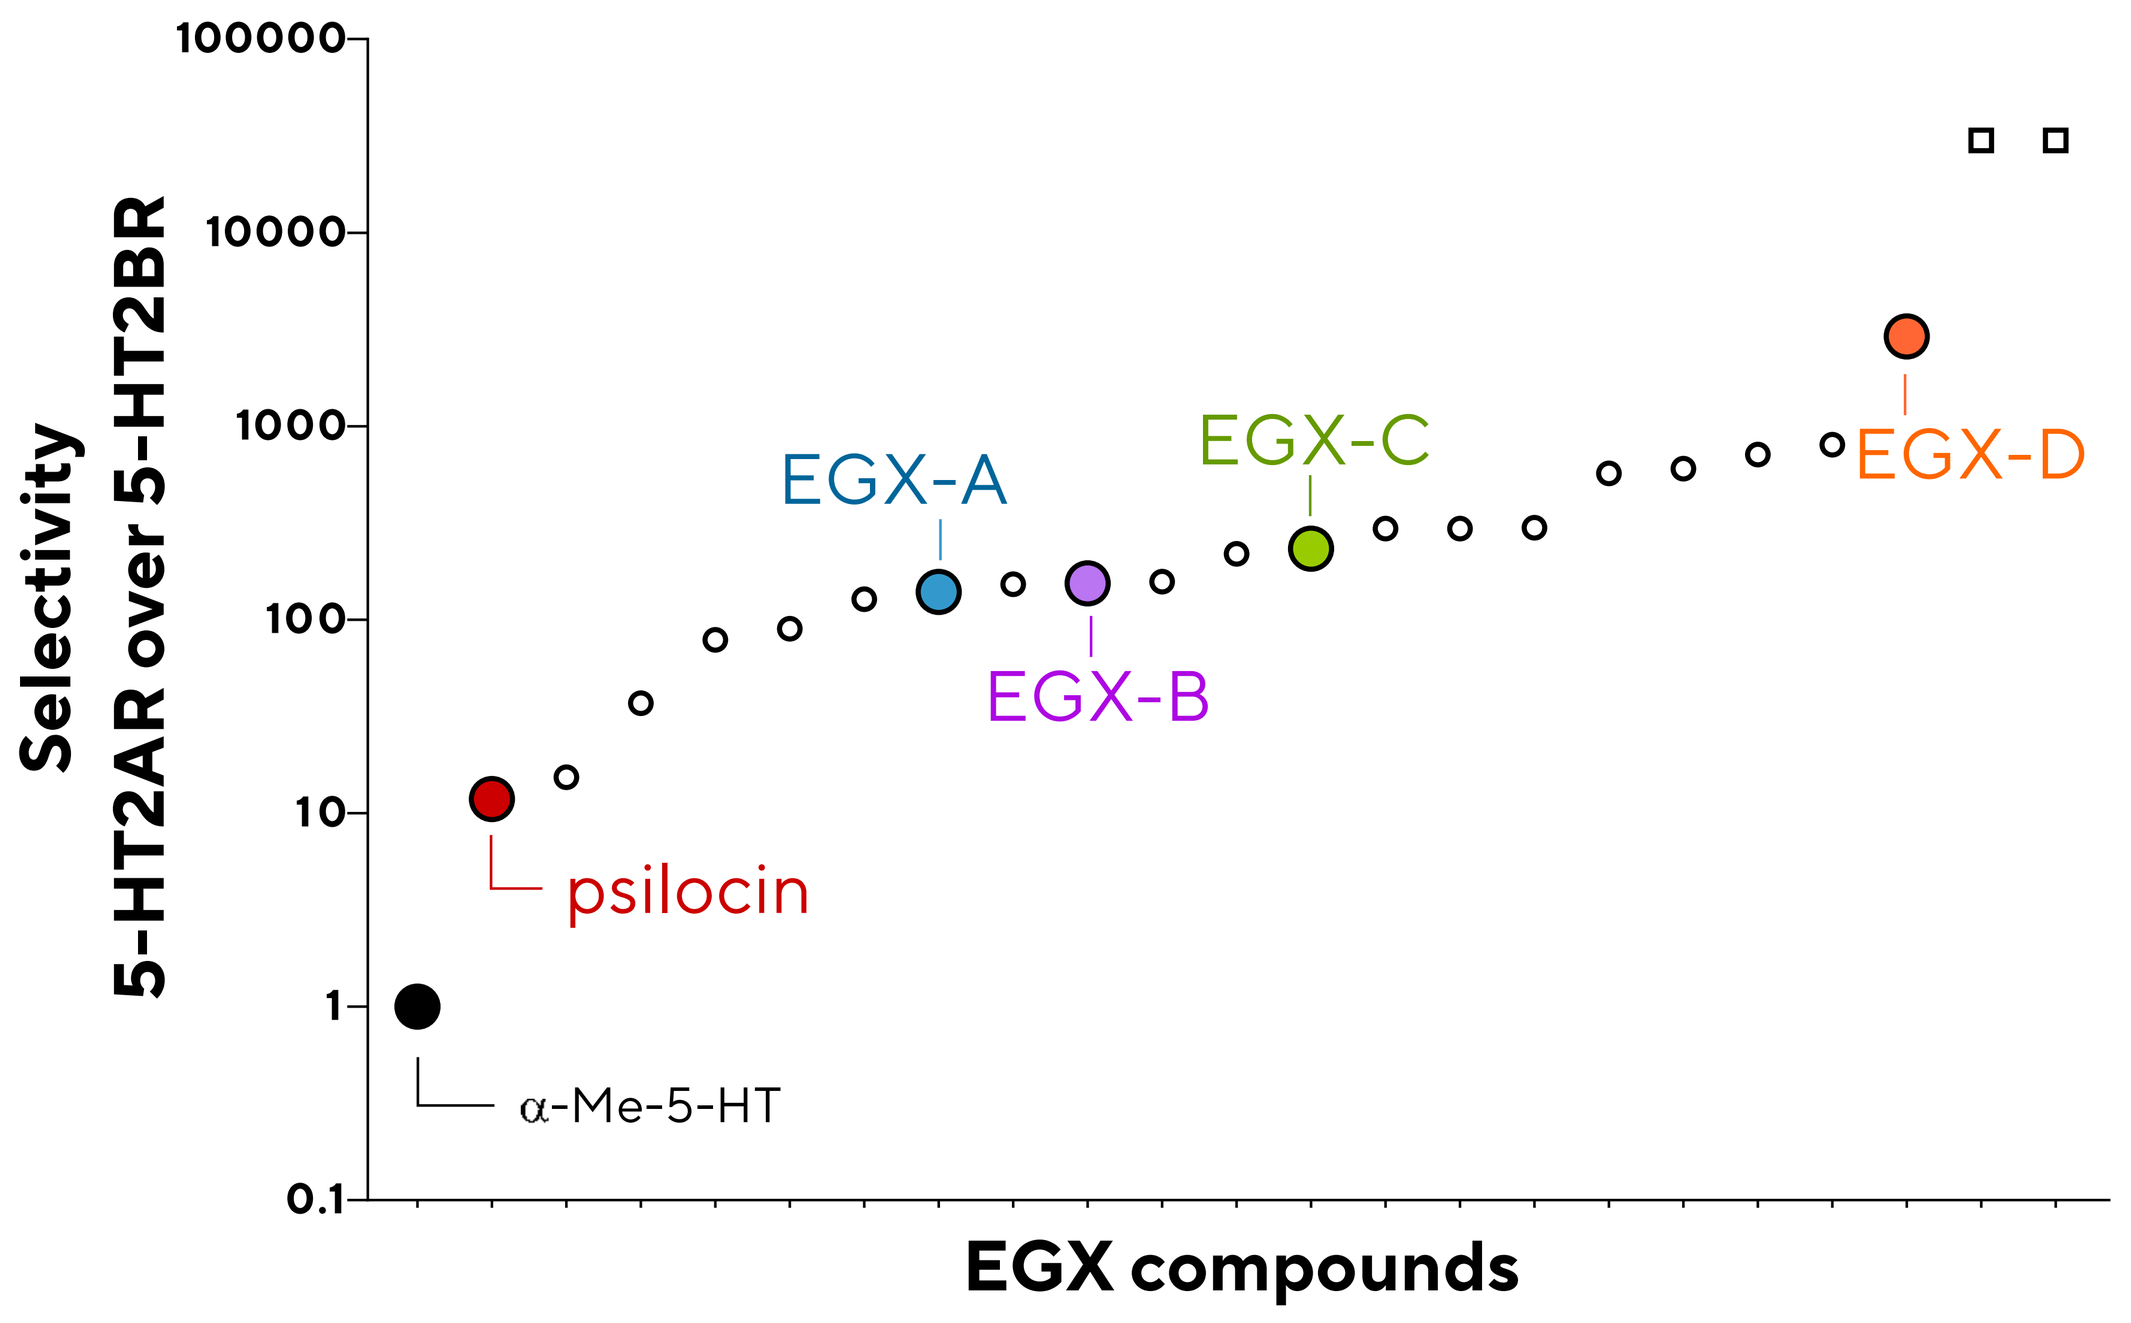
Selectivity
5-HT2AR over 5-HT2BR
100000
10000
1000
100
10
1
0.1
α-Me-5-HT
psilocin
EGX-A
EGX-B
EGX-C
EGX-D
EGX compounds

Figure 2. Novel EGX compounds rank-ordered by relative agonist selectivity for 5-HT2AR over 5-HT2BR (ΔΔlog(Emax/EC50); IPOne assay). Square symbols: compounds with selectivity ratios > 30,000. Assay reference agonist (α-Me-5-HT) used for normalization at each receptor (5-HT2AR/5-HT2BR selectivity = 1).

EGX-A Induced the HTR via 5-HT2AR Activation

Figure 3. EGX-A produced a significant dose-dependent increase in HTR counts in male C57BL/6J mice (ED_{50} = 1.7 mg/kg), an effect that was blocked by pretreatment with a 5-HT2AR antagonist, which is consistent with the reported effects of known psychedelic compounds.

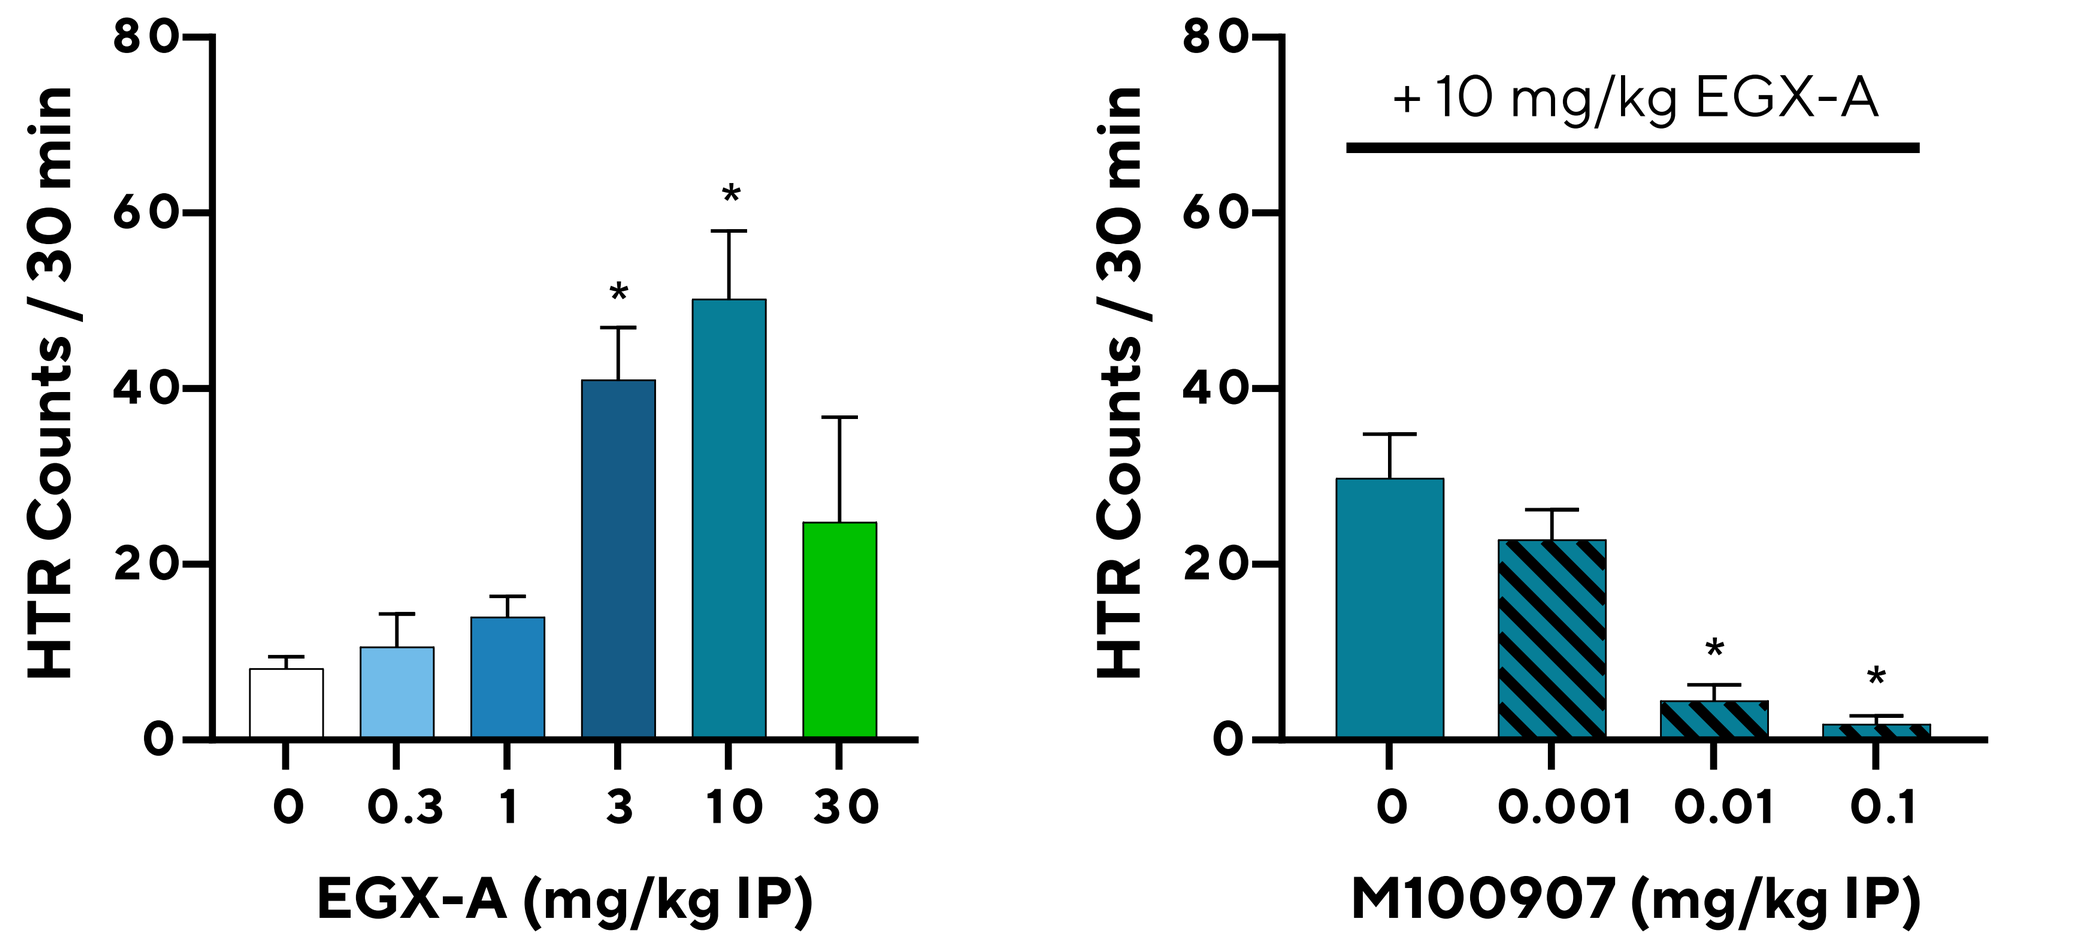
HTR Counts / 30 min
80
60
40
20
0
0 0.3 1 3 10 30
EGX-A (mg/kg IP)
HTR Counts / 30 min
+ 10 mg/kg EGX-A
0 0.001 0.01 0.1
M100907 (mg/kg IP)

Figure 3. HTR counts (mean ± SEM) over 30 min in mice following administration of EGX-A (0.3, 1, 3, 10, 30 mg/kg IP, left) or EGX-A (10 mg/kg IP) after 20 min pretreatment with the 5-HT2AR antagonist, M100907 (0.001, 0.01, 0.1 mg/kg IP, right). *p<0.05 vs. vehicle.

EGX-A Reduced Immobility in the Rat FST

Figure 4. EGX-A significantly reduced the immobility of male SD rats in the FST, similar to the rapid-acting antidepressant ketamine (Ket) and indicative of potential antidepressant drug-like activity in this screening assay.

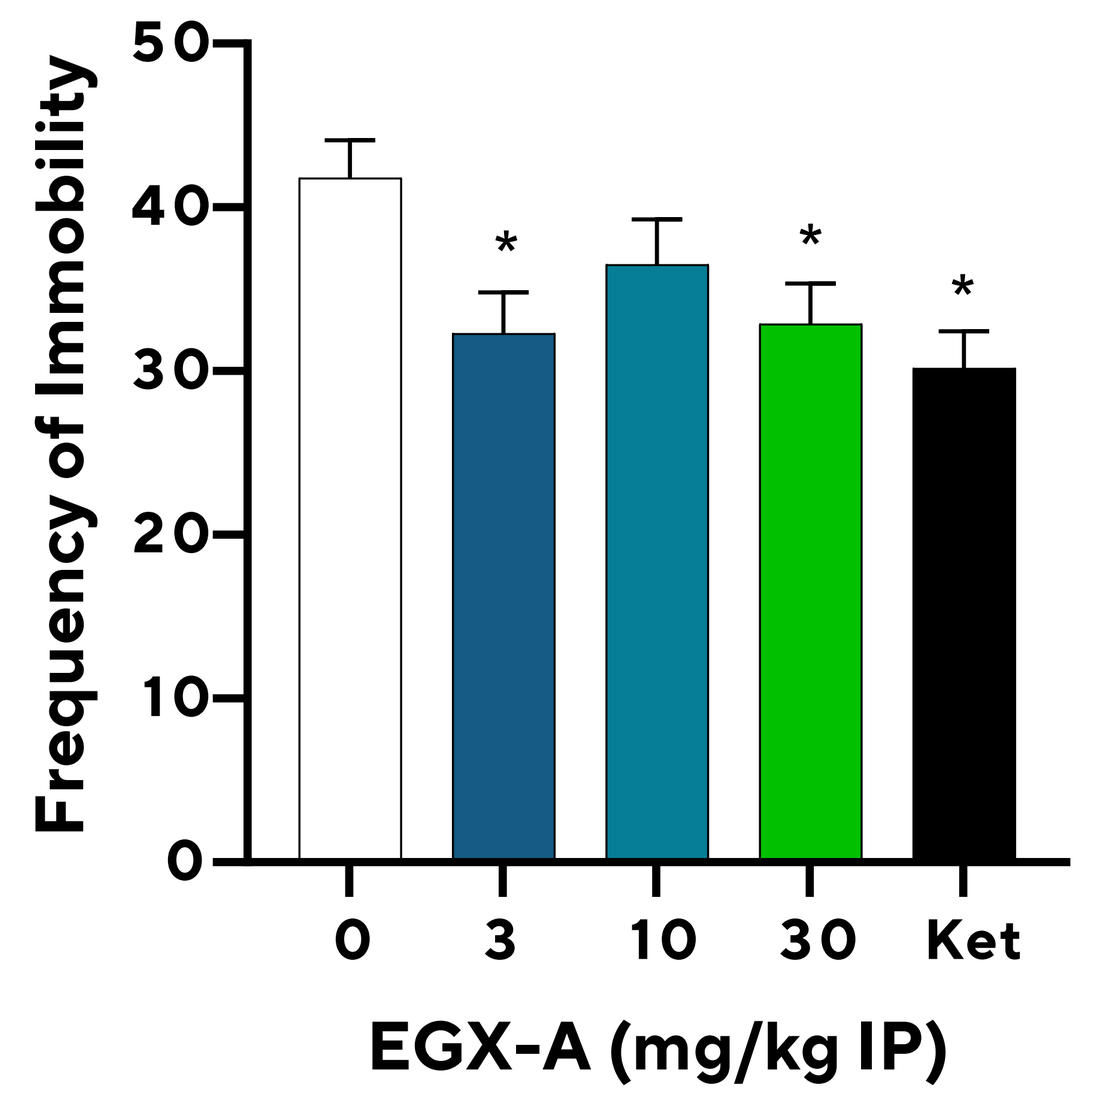
Frequency of Immobility
50
40
30
20
10
0
0 3 10 30 Ket
EGX-A (mg/kg IP)

Figure 4. Frequency of immobility (mean ± SEM) following administration of EGX-A (3, 10, 30 mg/kg IP) or ketamine (10 mg/kg IP) to male SD rats. *p<0.05 vs. vehicle. Five-minute tests were recorded, and video behavioral scoring was performed by trained technicians using a 5-second time-sampling technique.

EGX-A Reduced REM Sleep in the WKY Rat TRD Model

Figure 5. EGX-A produced a significant dose-dependent increase of REM sleep latency in male WKY rats, similar to the prototypical psychedelic psilocybin (Psi) and indicative of translational antidepressant drug-like activity.

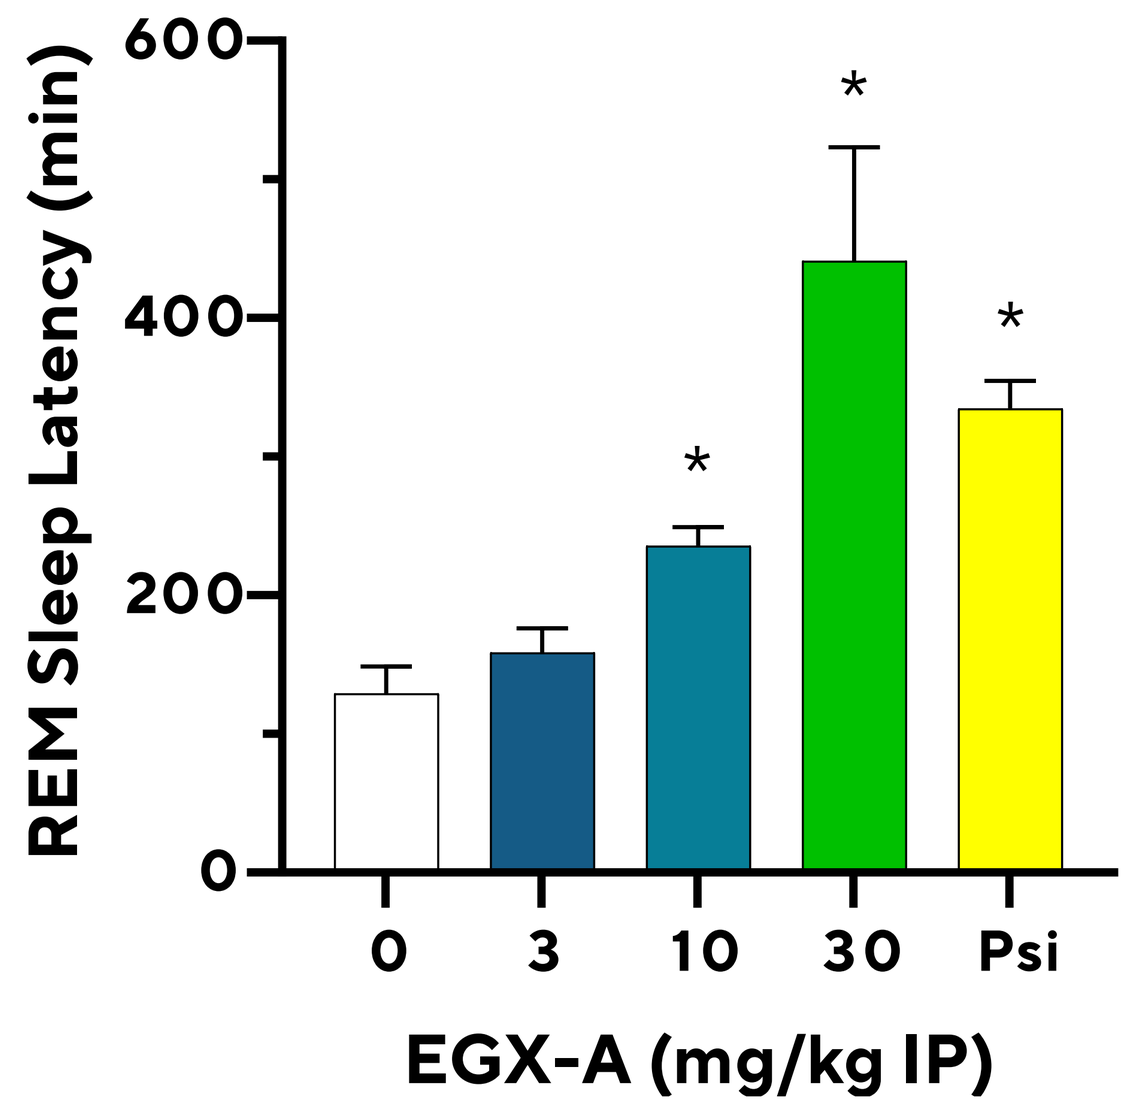
REM Sleep Latency (min)
600
400
200
0
0 3 10 30 Psi
EGX-A (mg/kg IP)

Figure 5. REM sleep latency (mean ± SEM) following administration of EGX-A (3, 10, 30 mg/kg IP) or psilocybin (10 mg/kg IP) to male WKY rats. *p<0.05 vs. vehicle.

Figure 6. EGX-A produced a significant dose-dependent decrease of REM sleep duration in male WKY rats, similar to psilocybin (Psi) and indicative of translational antidepressant drug-like activity in a model of treatment-resistant depression.

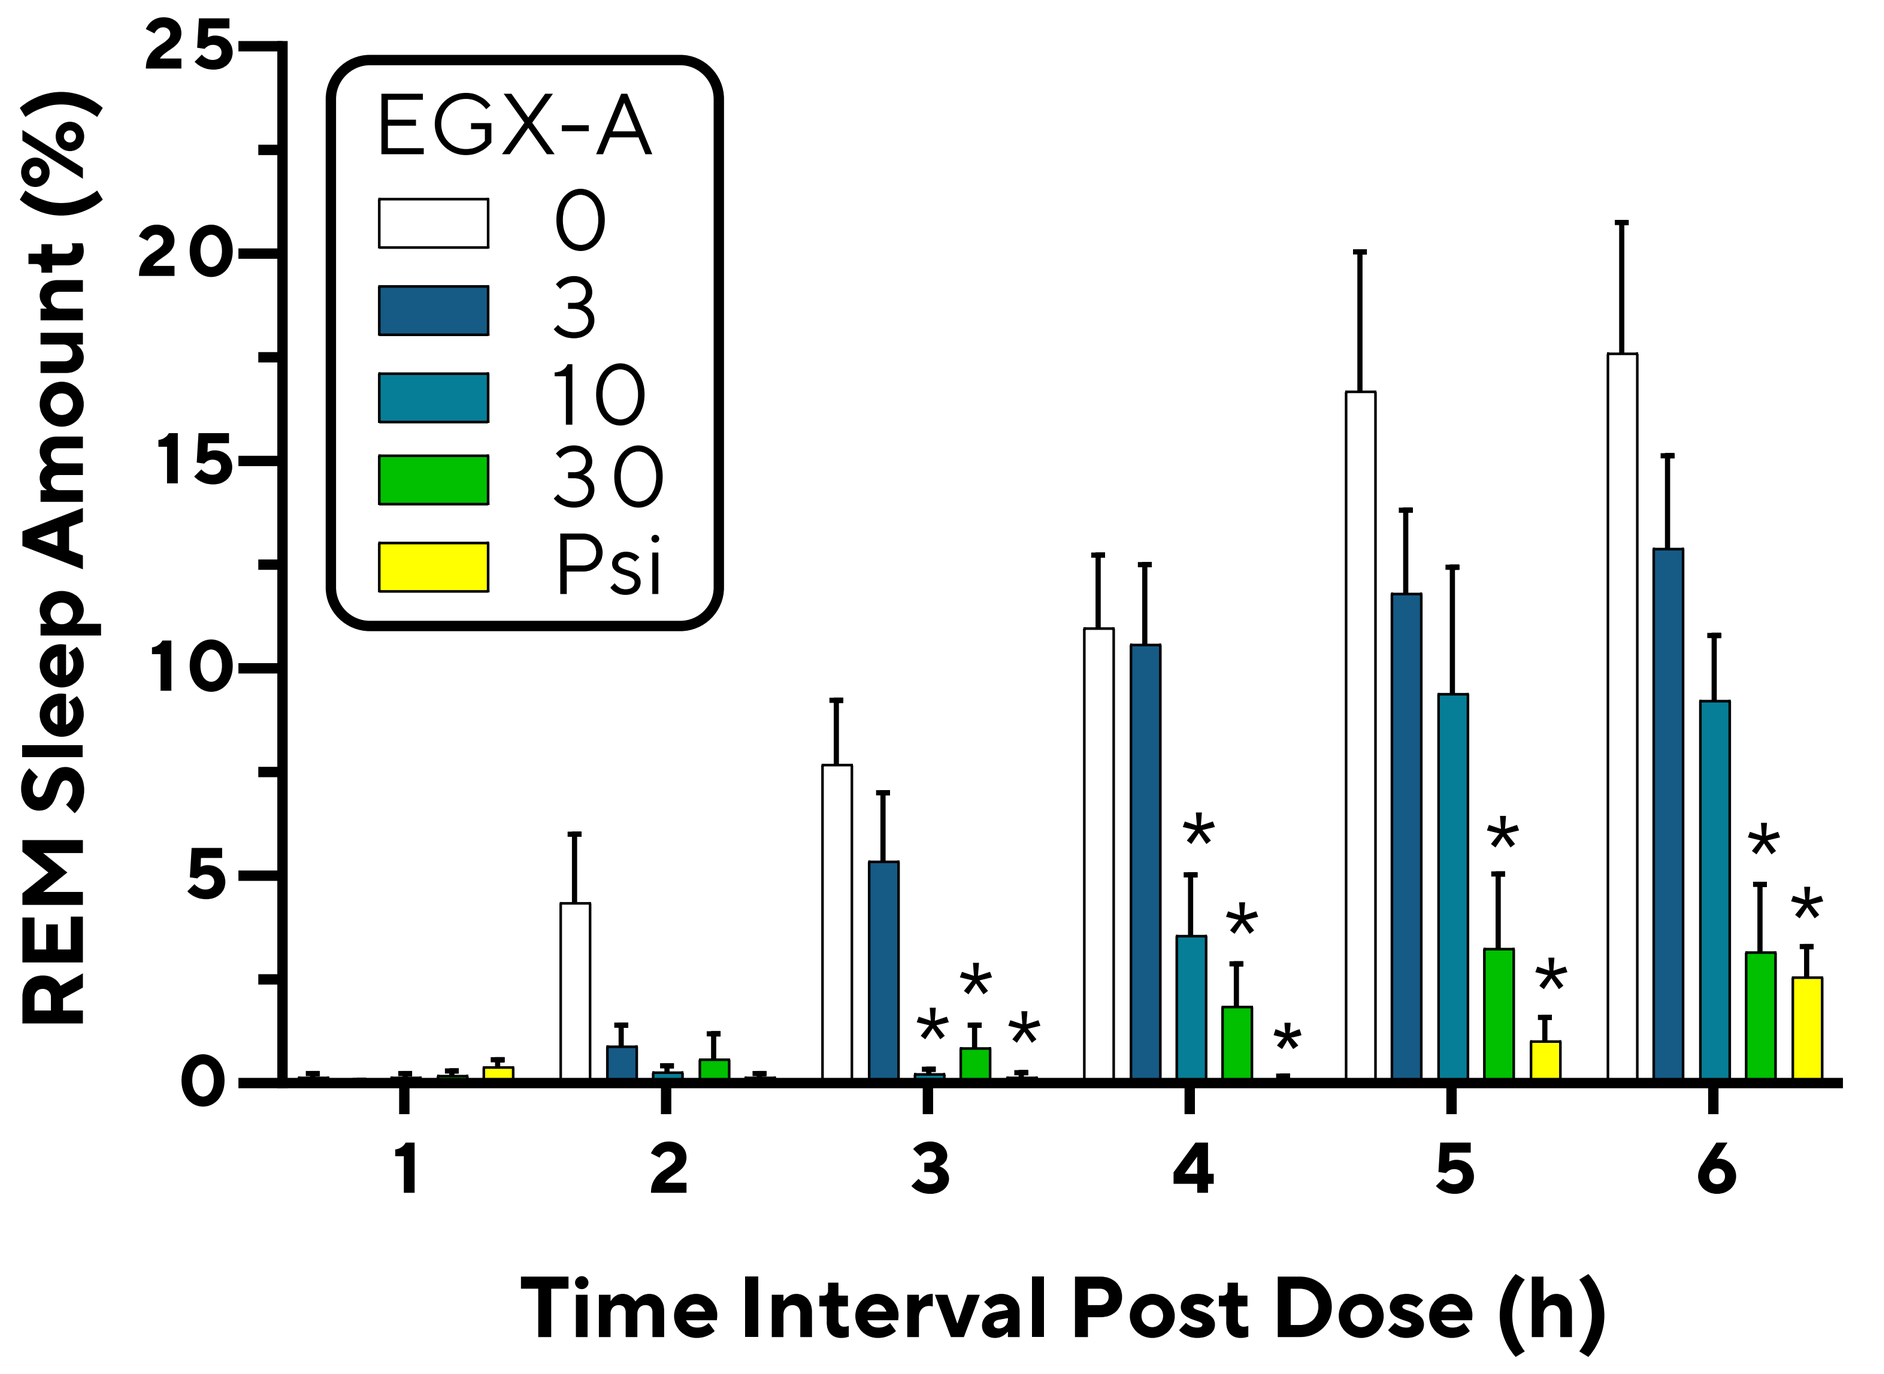
REM Sleep Amount (%)
25
20
15
10
5
0
EGX-A
0
3
10
30
Psi
1 2 3 4 5 6
Time Interval Post Dose (h)

Figure 6. Percentage of time in REM sleep (mean ± SEM) for a 6h assessment period following administration of EGX-A (3, 10, 30 mg/kg IP) or psilocybin (10 mg/kg IP) to male WKY rats. *p<0.05 vs. vehicle.

Summary and Conclusions

In Vitro Pharmacology

- Novel molecules were discovered that exhibited increased 5-HT2AR agonist activity, as well as enhanced selectivity for 5-HT2AR over 5-HT2BR relative to psilocin, the active metabolite of psilocybin
- EGX-A was identified as a potent 5-HT2AR full agonist with a high degree of selectivity for 5-HT2AR over 5-HT2BR

In Vivo Pharmacology

- **Mouse HTR:** EGX-A induced the HTR in a dose- and 5-HT2AR-dependent manner. As a strong positive correlation was reported between HTR activity in mice and hallucinogenic potential in humans [4], these results indicate that EGX-A may produce psychedelic effects in humans
- **SD Rat FST:** EGX-A reduced immobility in the FST, similar to the effect of the rapid-acting antidepressant ketamine, suggesting potential antidepressant drug-like activity
- **WKY rat REM Sleep:** EGX-A dose-dependently increased REM sleep latency and suppressed REM sleep duration in the WKY rat model of TRD, similar to the effects of the serotonergic psychedelic psilocybin. The EGX-A profile was consistent with reported effects of psychedelic 5-HT2AR agonists (e.g., psilocybin/psilocin, DOI) in healthy subjects and rodents [10-13] as well as acute and chronic treatment effects of several classes of antidepressant drugs in humans [6-7], supporting potential antidepressant drug-like activity in humans

Together, these results suggest that

- Novel agonists with selectivity for 5-HT2AR over 5-HT2BR may exhibit improved cardiac safety profiles relative to non-selective 5-HT2AR agonists
- One such novel 5-HT2AR agonist, EGX-A, shows evidence of psychedelic potential and antidepressant-like activity, based on positive data from preclinical assays with translational relevance

Acknowledgement: atai Life Sciences would like to thank all colleagues involved in the Entheogenix Biosciences program as well as the teams of external service providers that supported this project.

References: 1. Voineskos, D., et al. (2020). Neuropsychiatr Dis Treat 16, 221-234; 2. Goodwin, G. M., et al. (2022). N Engl J Med 387, 1637-1648; 3. Hibicke, M., et al. (2020). ACS Chem Neurosci 11, 864-871; 4. Halberstadt, A. L., et al. (2020). Neuropharmacology 167, 107933 https://doi.org/10.1016/j.neuropharm.2019.107933; 5. Palagini, L., et al. (2012). Sleep Med Rev 17(5), 377-390; 6. Wichniak, A., et al. (2017). Curr Psychiatry Rep 19, 63 https://doi.org/10.1007/s11920-017-0816-4; 7. Wilson, S., et al. (2005). Drugs 65(7), 927-947; 8. Ivarsson, M., et al. (2005). Eur J Pharmacol 522, 63-71; 9. Halberstadt, A. L. (2020). Sci Rep 10, 8344 doi: 10.1038/s41598-020-65264-x; 10. Dudysova, D., et al. (2020). Front Pharmacol 11:602590. doi: 10.3389/fphar.2020.602590; 11. Monti, J. M., et al. (2006). Eur J Pharmacol 55(1-3), 163-170; 12. Thomas, C., et al. (2020). Neuropsychopharmacology 45(SUPPL 1), 138-139; 13. Thomas, C. W., et al. (2022). Translational Psychiatry 12, 77 https://doi.org/10.1038/s41398-022-01846-9

Author Disclosure Information: C.A. Bowen: Employment/Salary (full or part-time): Employee of atai Life Sciences. T.A. Khan: Employment/Salary (full or part-time): Employee of atai Life Sciences. G.F. Short, III: Employment/Salary (full or part-time): Employee of atai Life Sciences. S. Rao: Employment/Salary (full or part-time): Employee of atai Life Sciences. P. Kleine: Employment/Salary (full or part-time): Employee of atai Life Sciences. A. Gibbs: Employment/Salary (full or part-time): Former Employee of atai Life Sciences. R. Aron: Employment/Salary (full or part-time): Former Employee of atai Life Sciences. R. Shin: Employment/Salary (full or part-time): Former Employee of atai Life Sciences. I. Premoli: Employment/Salary (full or part-time): Former Employee of atai Life Sciences. R.B. Perni: Consultant/Service Provider for atai Life Sciences. A.L. Halberstadt: Consultant/Service Provider for atai Life Sciences.